Targeting of a cancer-associated LYPD3 glycoform for tumor therapy

Evelyn Hartung, Antje Danielczyk, Johanna Gellert, Stephanie Gurka, Anika Jäkel, Naomi Kast, Timo Lischke, Theresa Neumann, Manon Weiske, Lisa Weiß, Luisa Willmann, Patrik Kehler

Correspondence to **Evelyn.Hartung@glycotope.com;** Glycotope, Berlin, Germany

Abstract 1347

Introduction

- In cancer therapy, normal tissue abundance of a target antigen may cause unwanted side effects using classical protein binding antibodies that cannot discriminate between the cancer-associated and healthy tissue target antigen
- Glycosylation is strongly altered in cancer, reflecting drastic changes in tumor metabolism [1] or genetic alterations
- Changes in tumor glycosylation mostly due to mutated or mislocated glycosyl-transferases and glycosidases give rise to **truncated O-glycans** [2] that are almost absent in healthy tissues:
 - **Thomsen nouvelle (Tn) antigen**: GalNAcα1-
 - **Sialylated Thomsen nouvelle (sTn) antigen sTn**: Neu5Ac(α2-6)GalNAcα1-
 - **Thomsen-Friedenreich (TF) antigen**: Gal(β1-3)GalNAcα1-

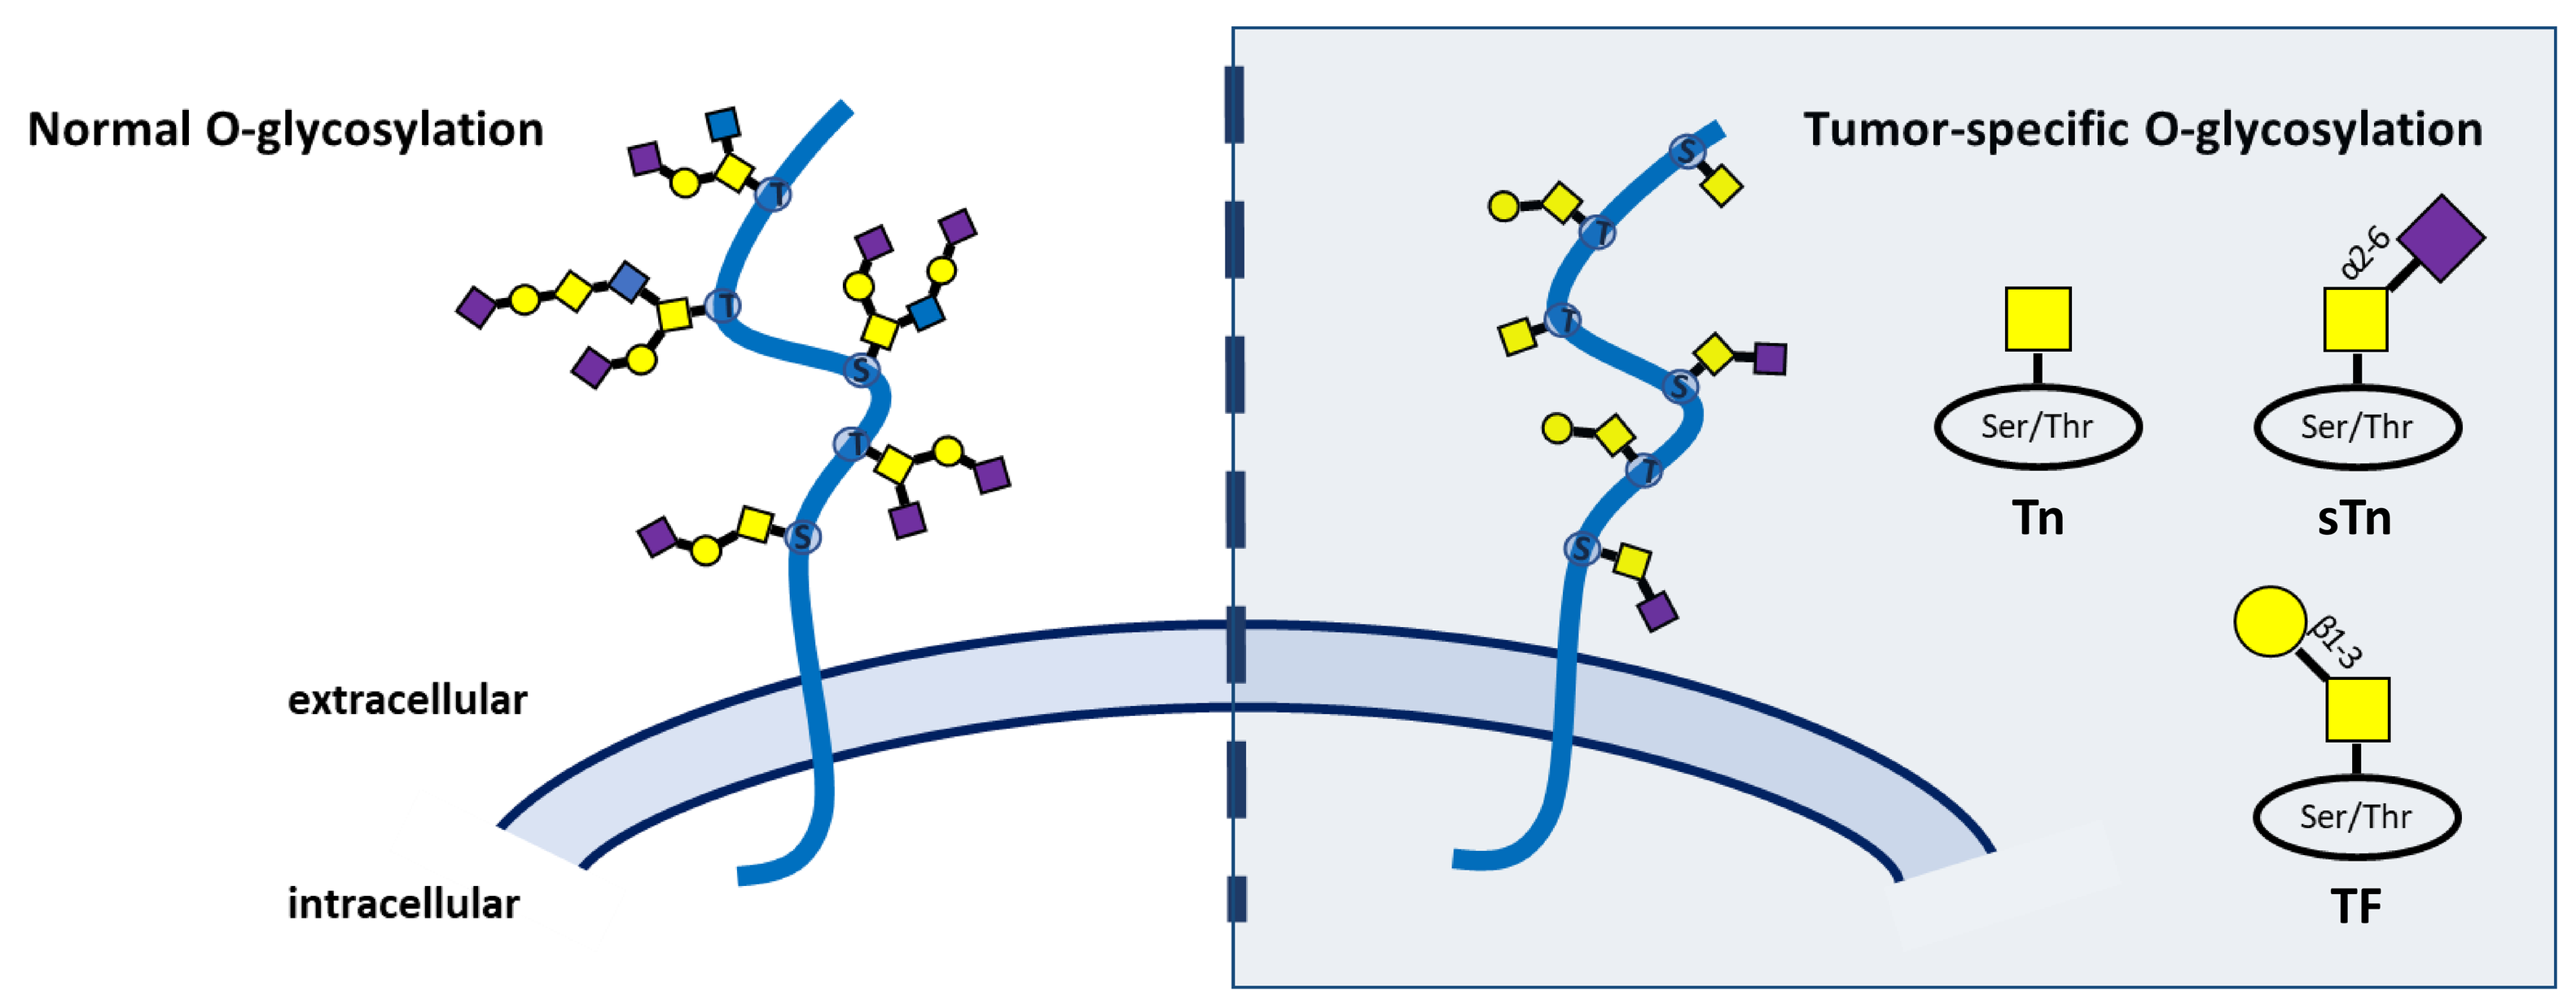
Normal O-glycosylation
Tumor-specific O-glycosylation
extracellular
intracellular
Ser/Thr
Tn
α2-6
sTn
β1-3
TF

- **LYPD3 (C4.4A)** is a glycosylphosphatidylinositol (GPI)-anchored, **highly glycosylated cell surface protein that is associated with carcinogenesis** in several different squamous cell carcinoma (SCC) of the head and neck and esophageal SCC (ESCC) [3]
- Under normal physiologic conditions, LYPD3 is expressed in different epithelia and skin keratinocytes [4]
- We have developed an **anti-LYPD3 antibody (GT-002)** that targets the cancer-associated LYPD3 glycoform and thereby shows **superior tumor specificity** compared to conventional protein-binding anti-LYPD3 antibodies

Unique binding profile of GT-002

GT-002 specifically binds to O-glycosylated LYPD3 (ELISA, flow cytometry)

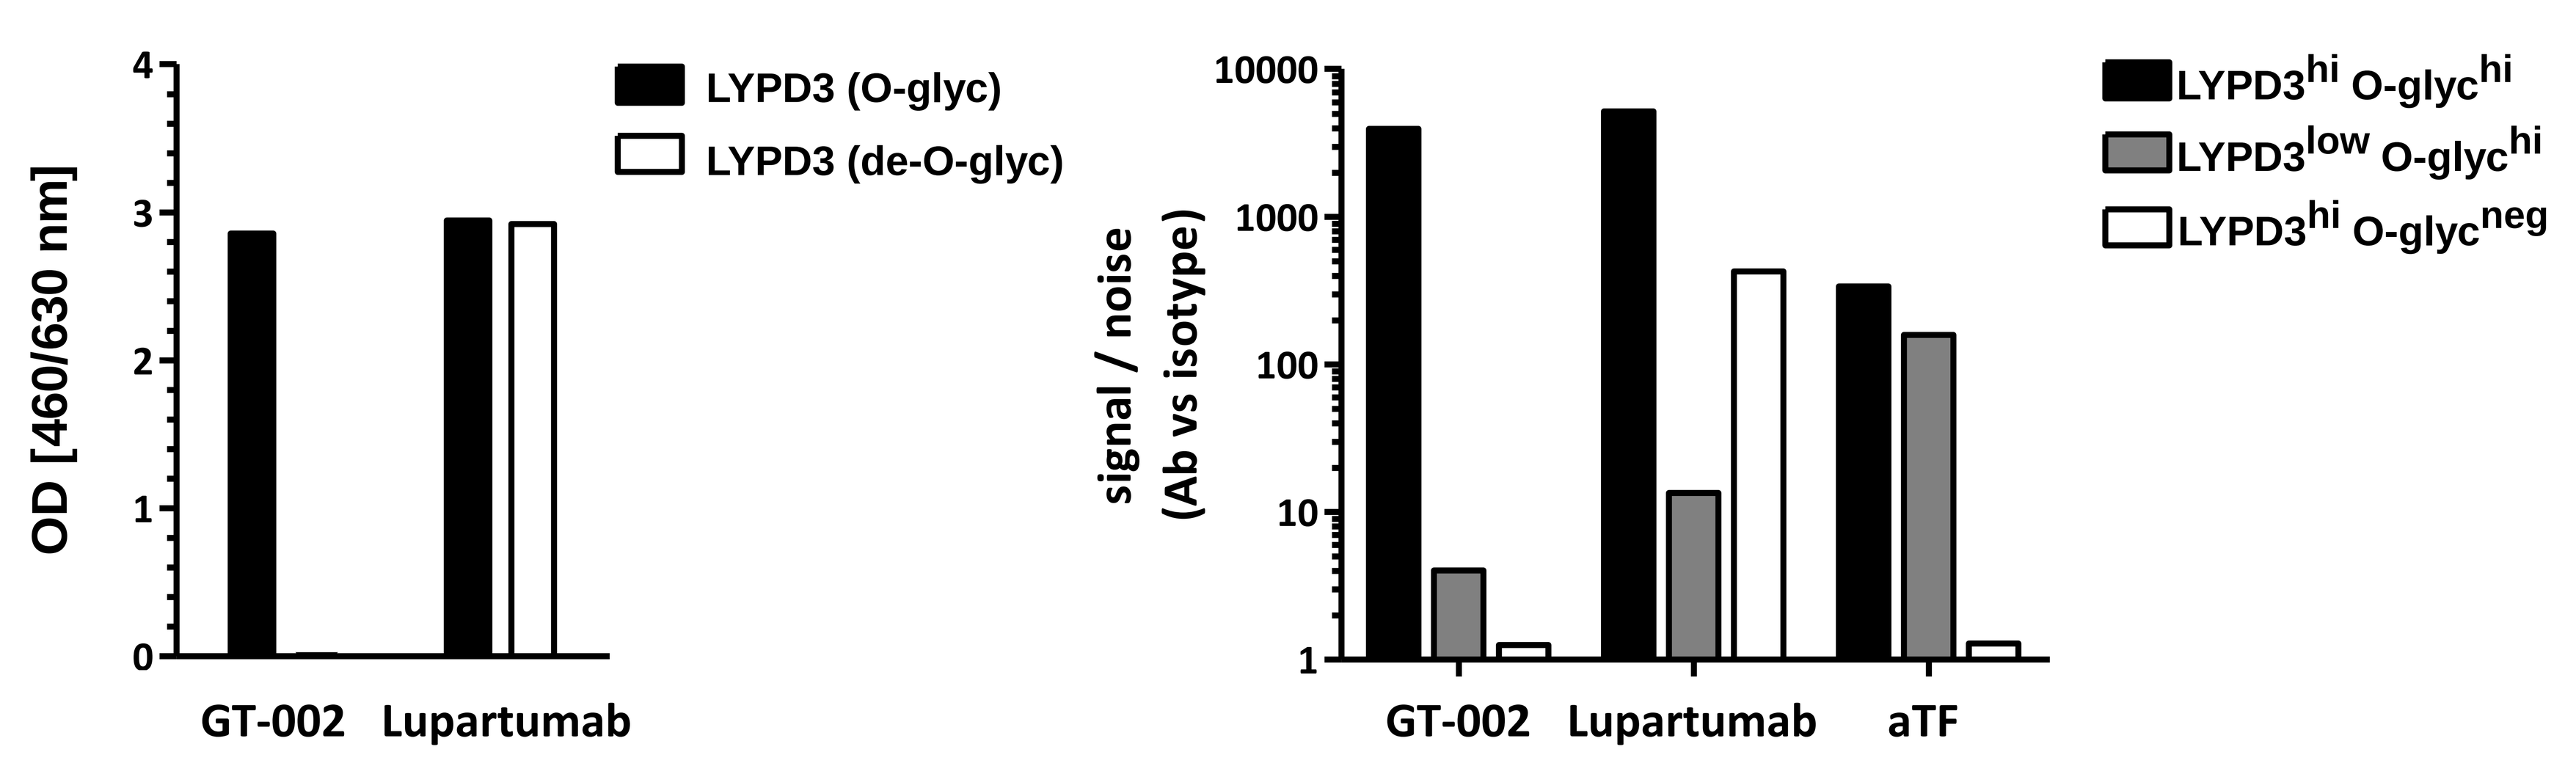
OD [460/630 nm]
LYPD3 (O-glyc)
LYPD3 (de-O-glyc)
GT-002
Lupartumab
signal / noise (Ab vs isotype)
LYPD3hi O-glychi
LYPD3low O-glychi
LYPD3hi O-glycneg
GT-002
Lupartumab
aTF

- Binding of GT-002 is highly restricted to O-glycosylated LYPD3 as there is no binding to de-O-glycosylated LYPD3 protein or to cells expressing LYPD3 without O-glycans
- A competitor - Lupartumab (BAY1112623) - recognizes LYPD3 independently of its glycosylation status

Unique binding profile of GT-002

GT-002 selectively binds to TF- but not Tn-carrying LYPD3 (ELISA)

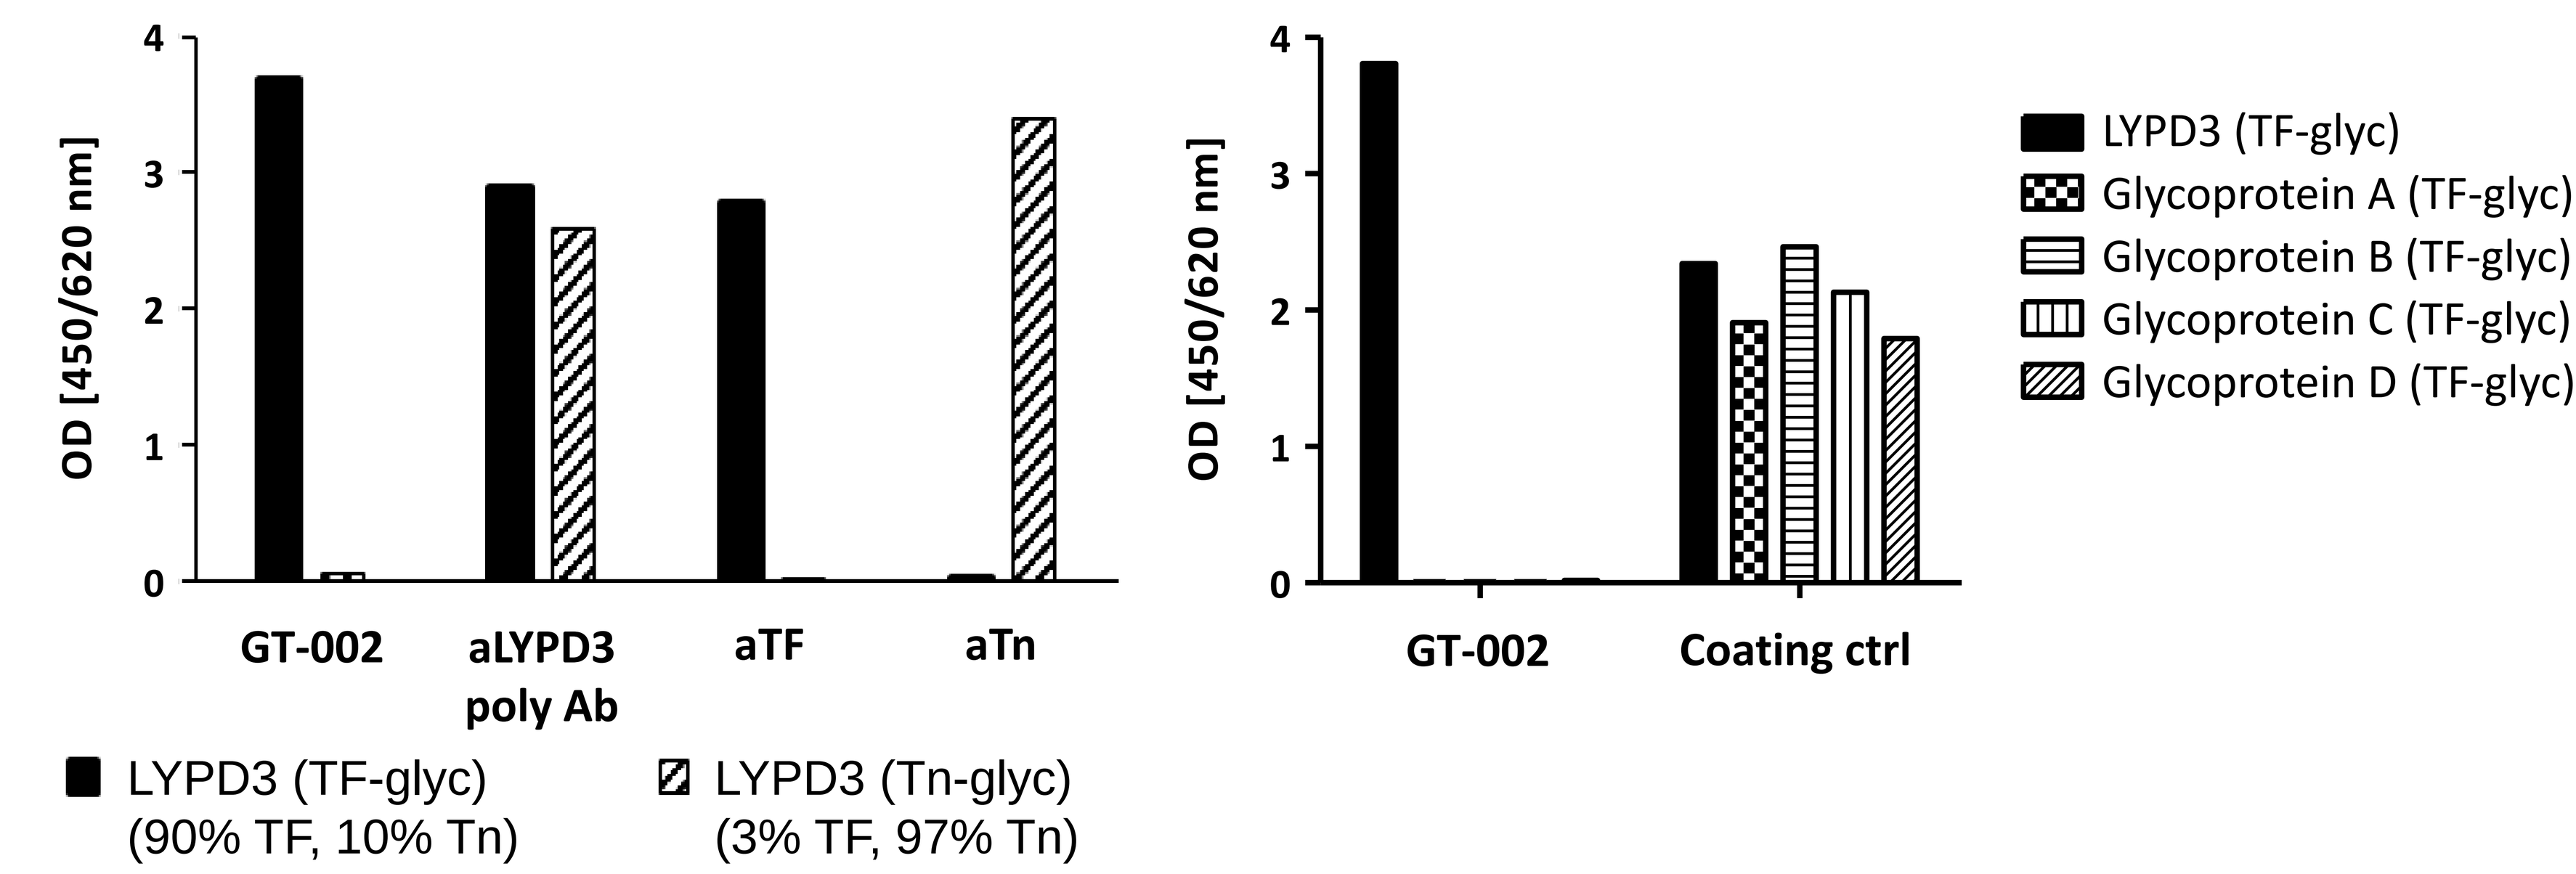
OD [450/620 nm]
GT-002
aLYPD3 poly Ab
aTF
aTn
LYPD3 (TF-glyc) (90% TF, 10% Tn)
LYPD3 (Tn-glyc) (3% TF, 97% Tn)
GT-002
Coating ctrl
LYPD3 (TF-glyc)
Glycoprotein A (TF-glyc)
Glycoprotein B (TF-glyc)
Glycoprotein C (TF-glyc)
Glycoprotein D (TF-glyc)

- The aLYPD3 poly Ab recognizes LYPD3 independently of its glycosylation status
- GT-002 binds only to TF-decorated LYPD3 but not to the closely related glycoprotein Tn-LYPD3 or other irrelevant glycoproteins harboring truncated O-glycans

GT-002 binds non-sialylated LYPD3 on tumor cell lines (flow cytometry)

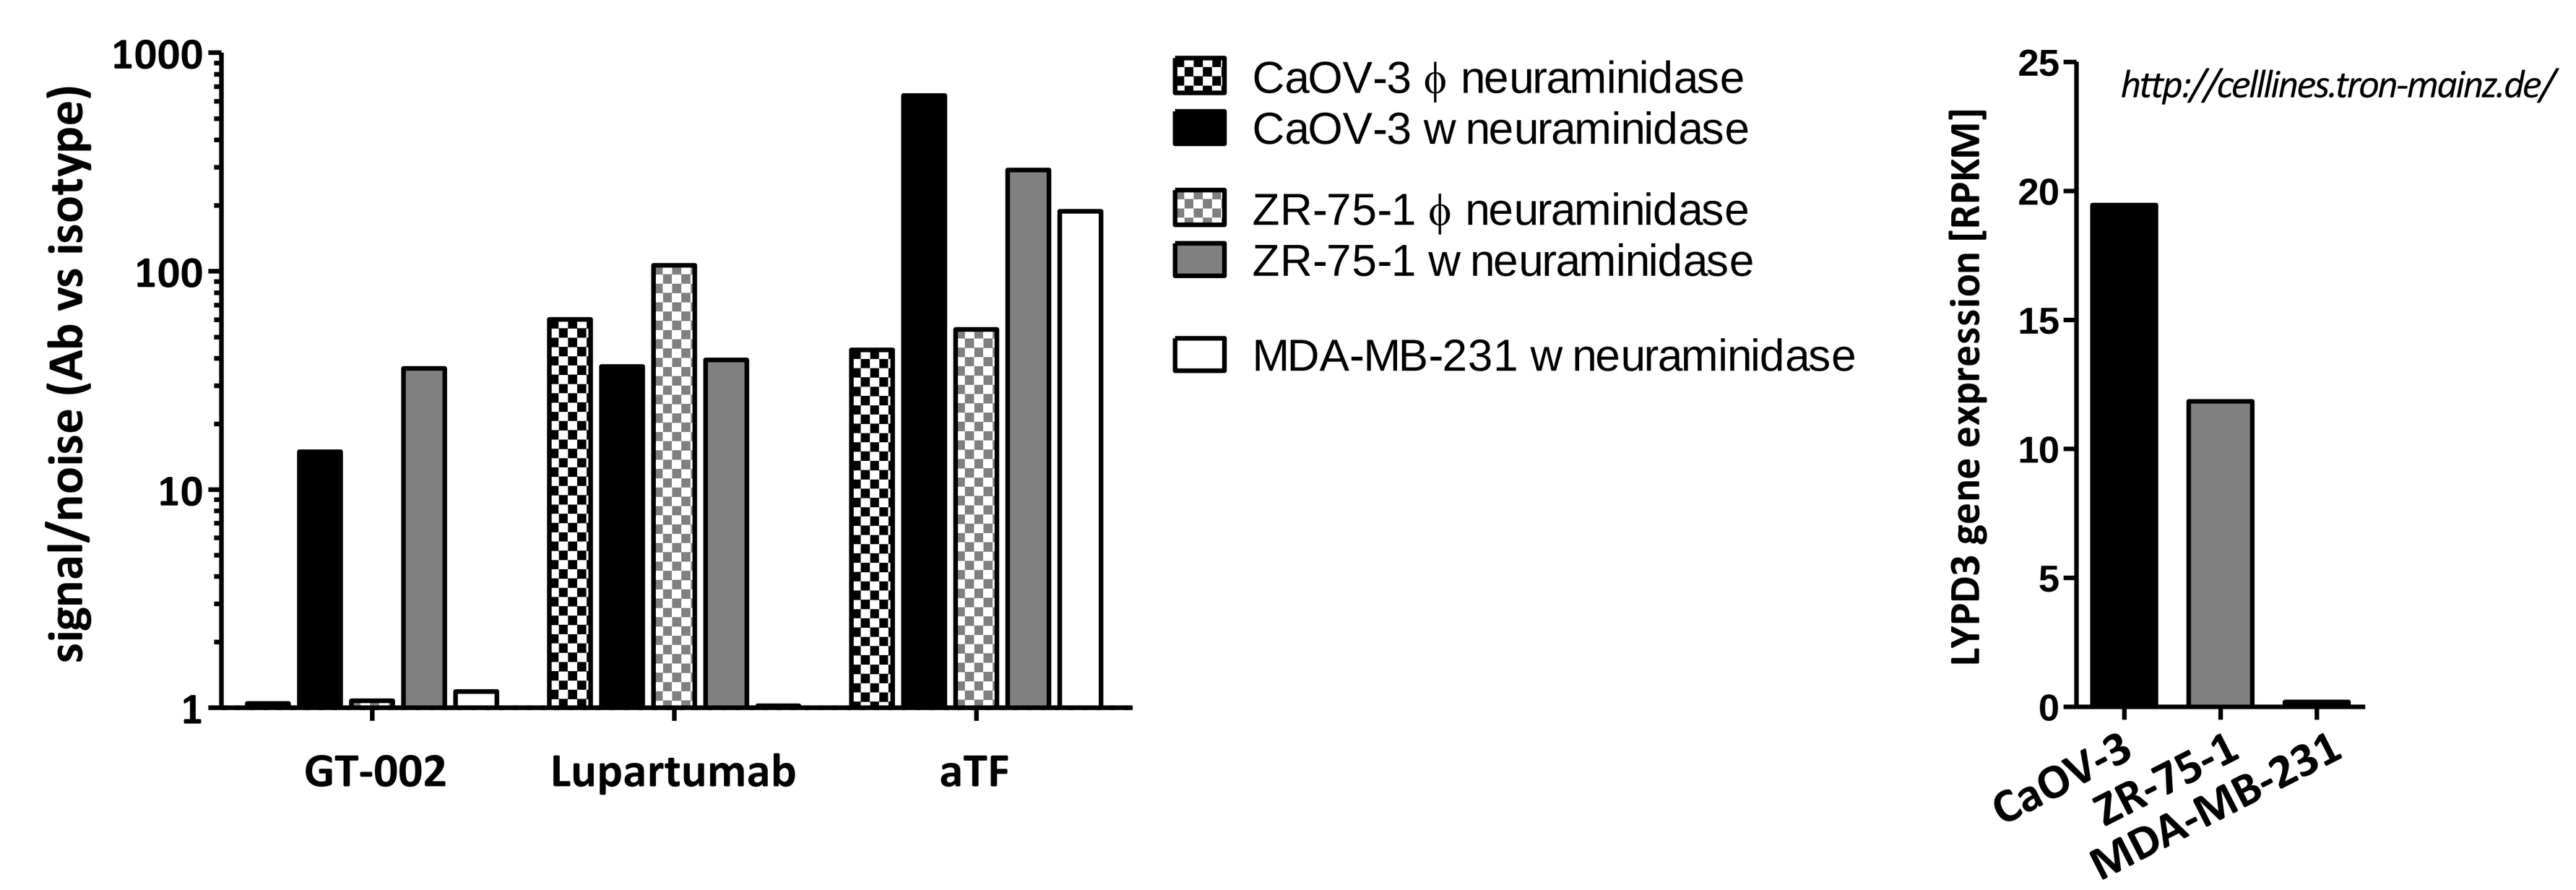
signal/noise (Ab vs isotype)
GT-002
Lupartumab
aTF
CaOV-3 ϕ neuraminidase
CaOV-3 w neuraminidase
ZR-75-1 ϕ neuraminidase
ZR-75-1 w neuraminidase
MDA-MB-231 w neuraminidase
LYPD3 gene expression [RPKM]
http://celllines.tron-mainz.de/
CaOV-3
ZR-75-1
MDA-MB-231

- Neuraminidase treatment is a robust method to release TF/Tn-LYPD3 glyco-epitopes on hypersialylating tumor cell lines in culture
- Binding pattern of GT-002 indicates strongly reduced binding to sialylated epitopes

Strongly reduced binding to normal tissue

Immunohistochemistry

H-Score (0-300) =
3 x percentage of strongly stained cells
+ 2 x percentage of moderately stained cells
+ percentage of weakly stained cells

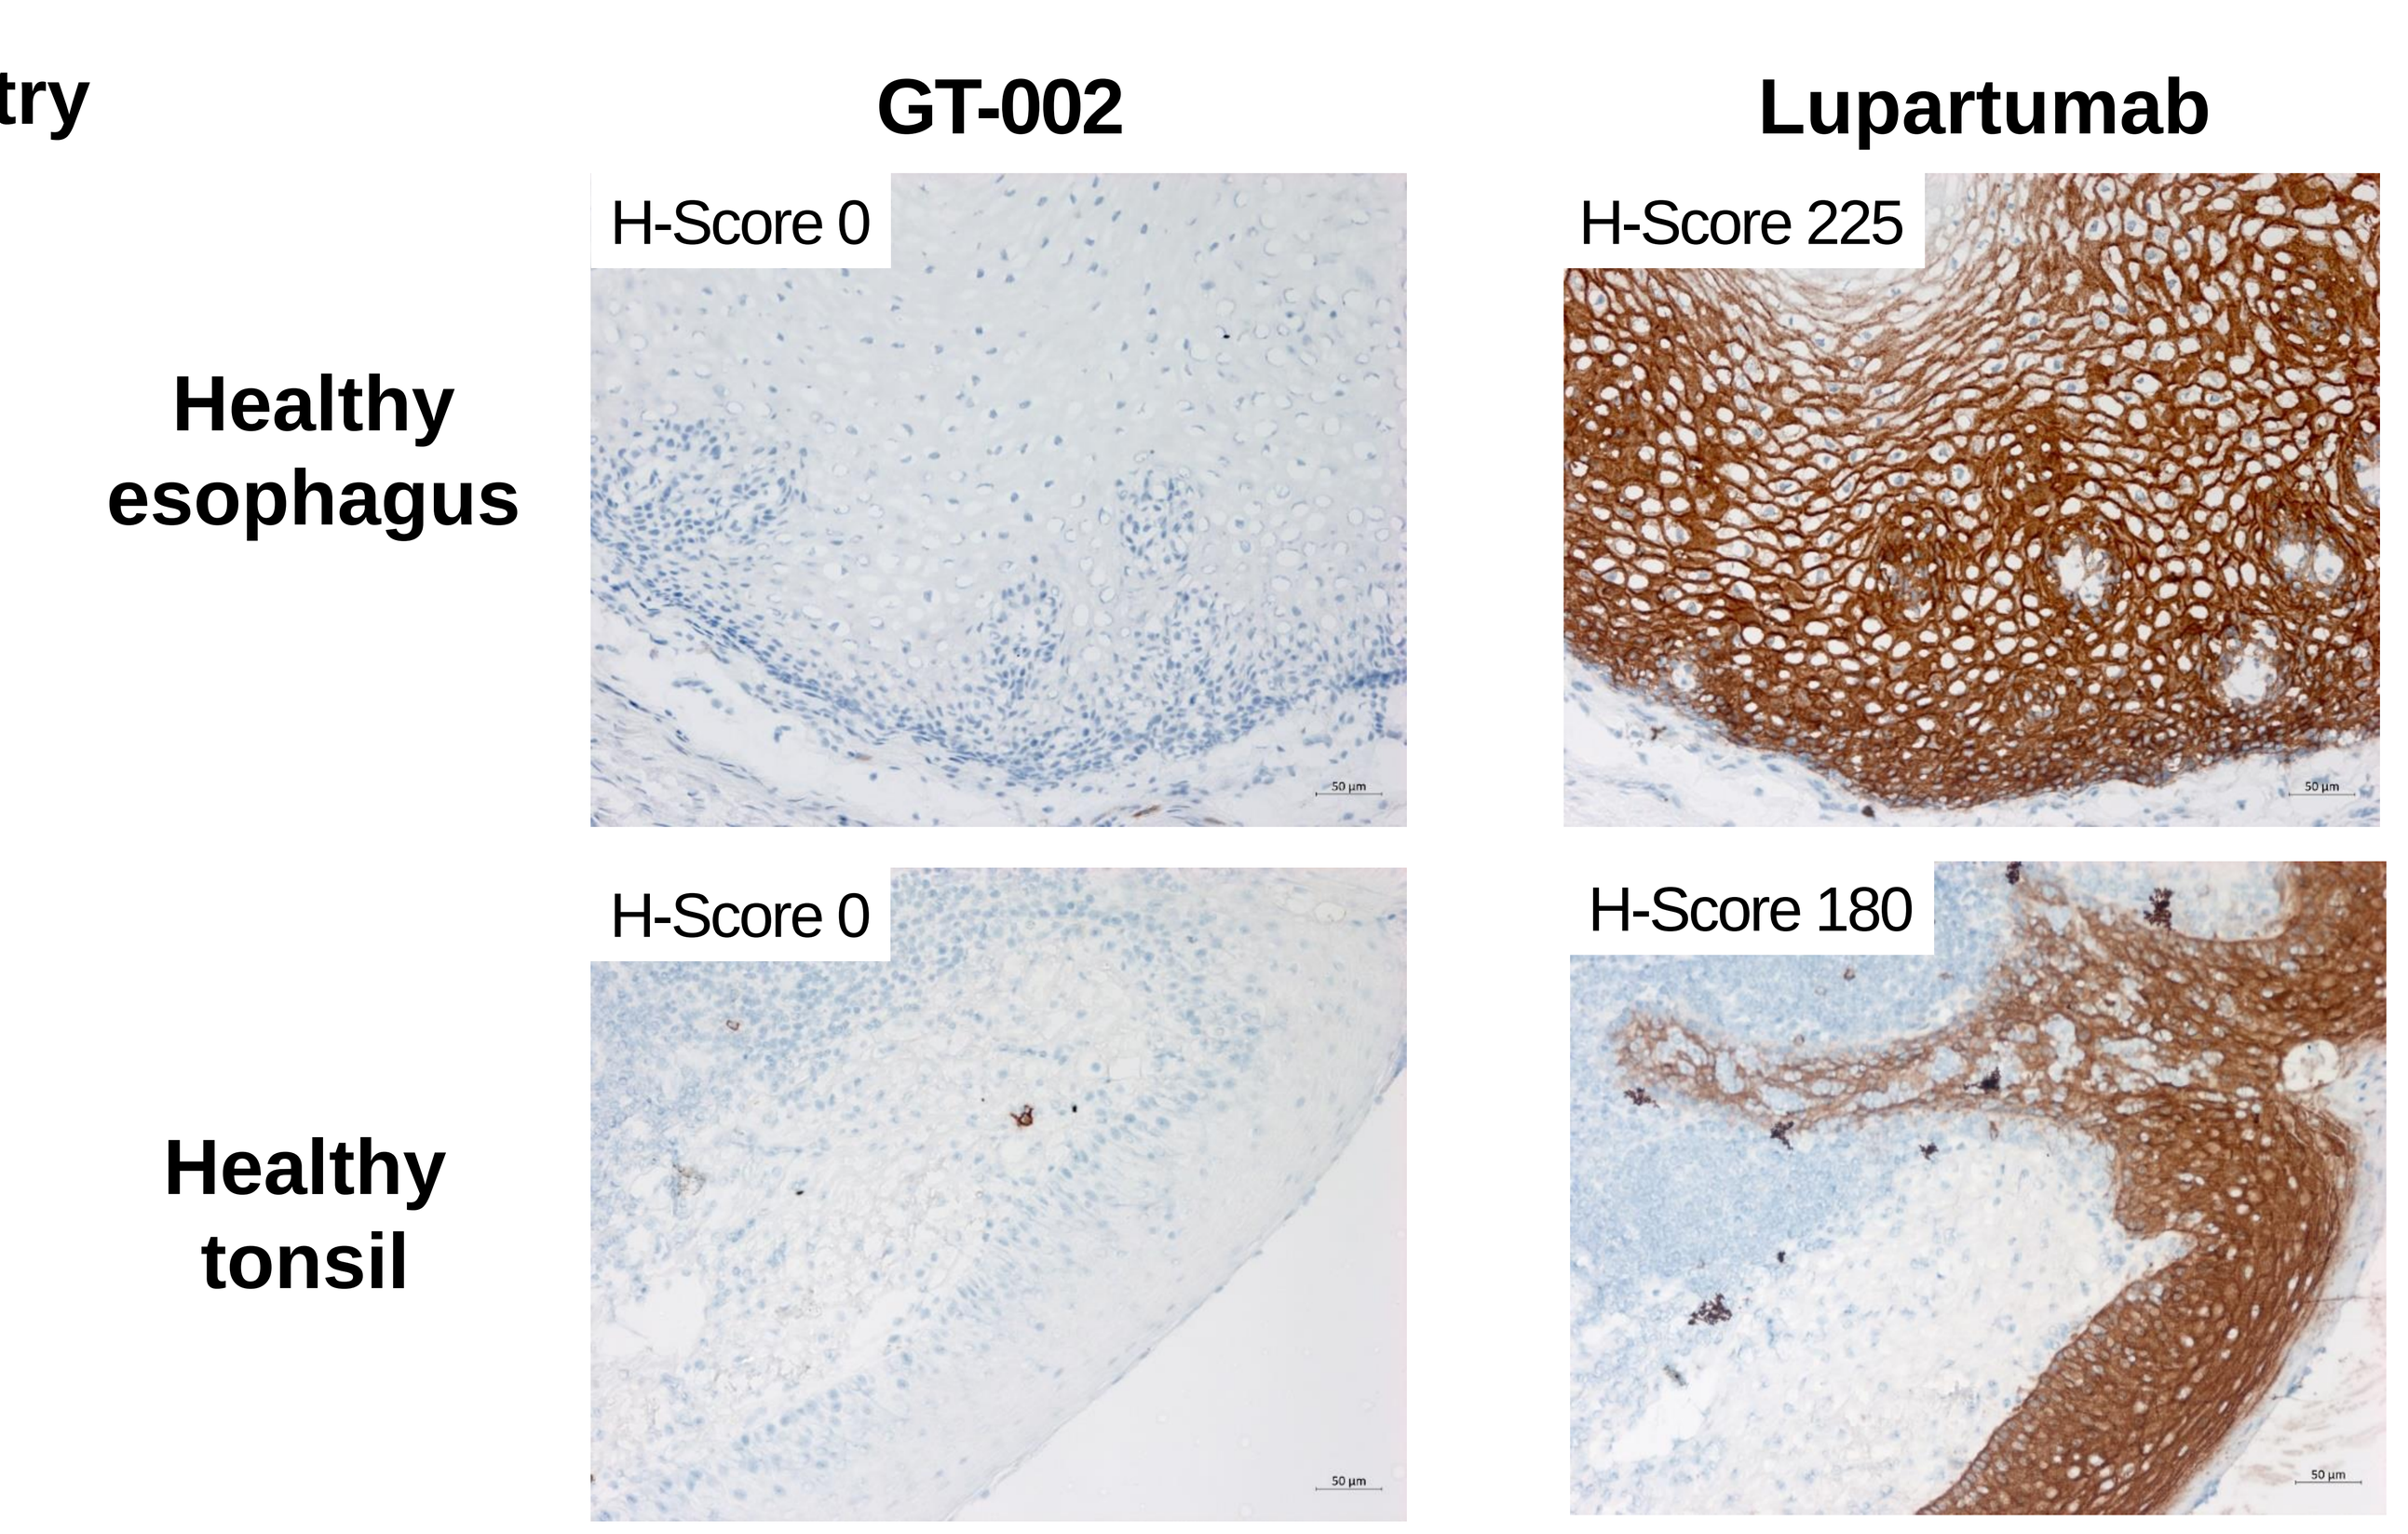
GT-002
Lupartumab
Healthy esophagus
H-Score 0
H-Score 225
Healthy tonsil
H-Score 0
H-Score 180

- Competitor Lupartumab binds to LYPD3 expressed in various healthy squamous epithelia
- GT-002 shows strongly reduced binding to LYPD3-positive squamous epithelia in healthy tissue

Selective recognition of SCC tumor tissue

Immunohistochemistry

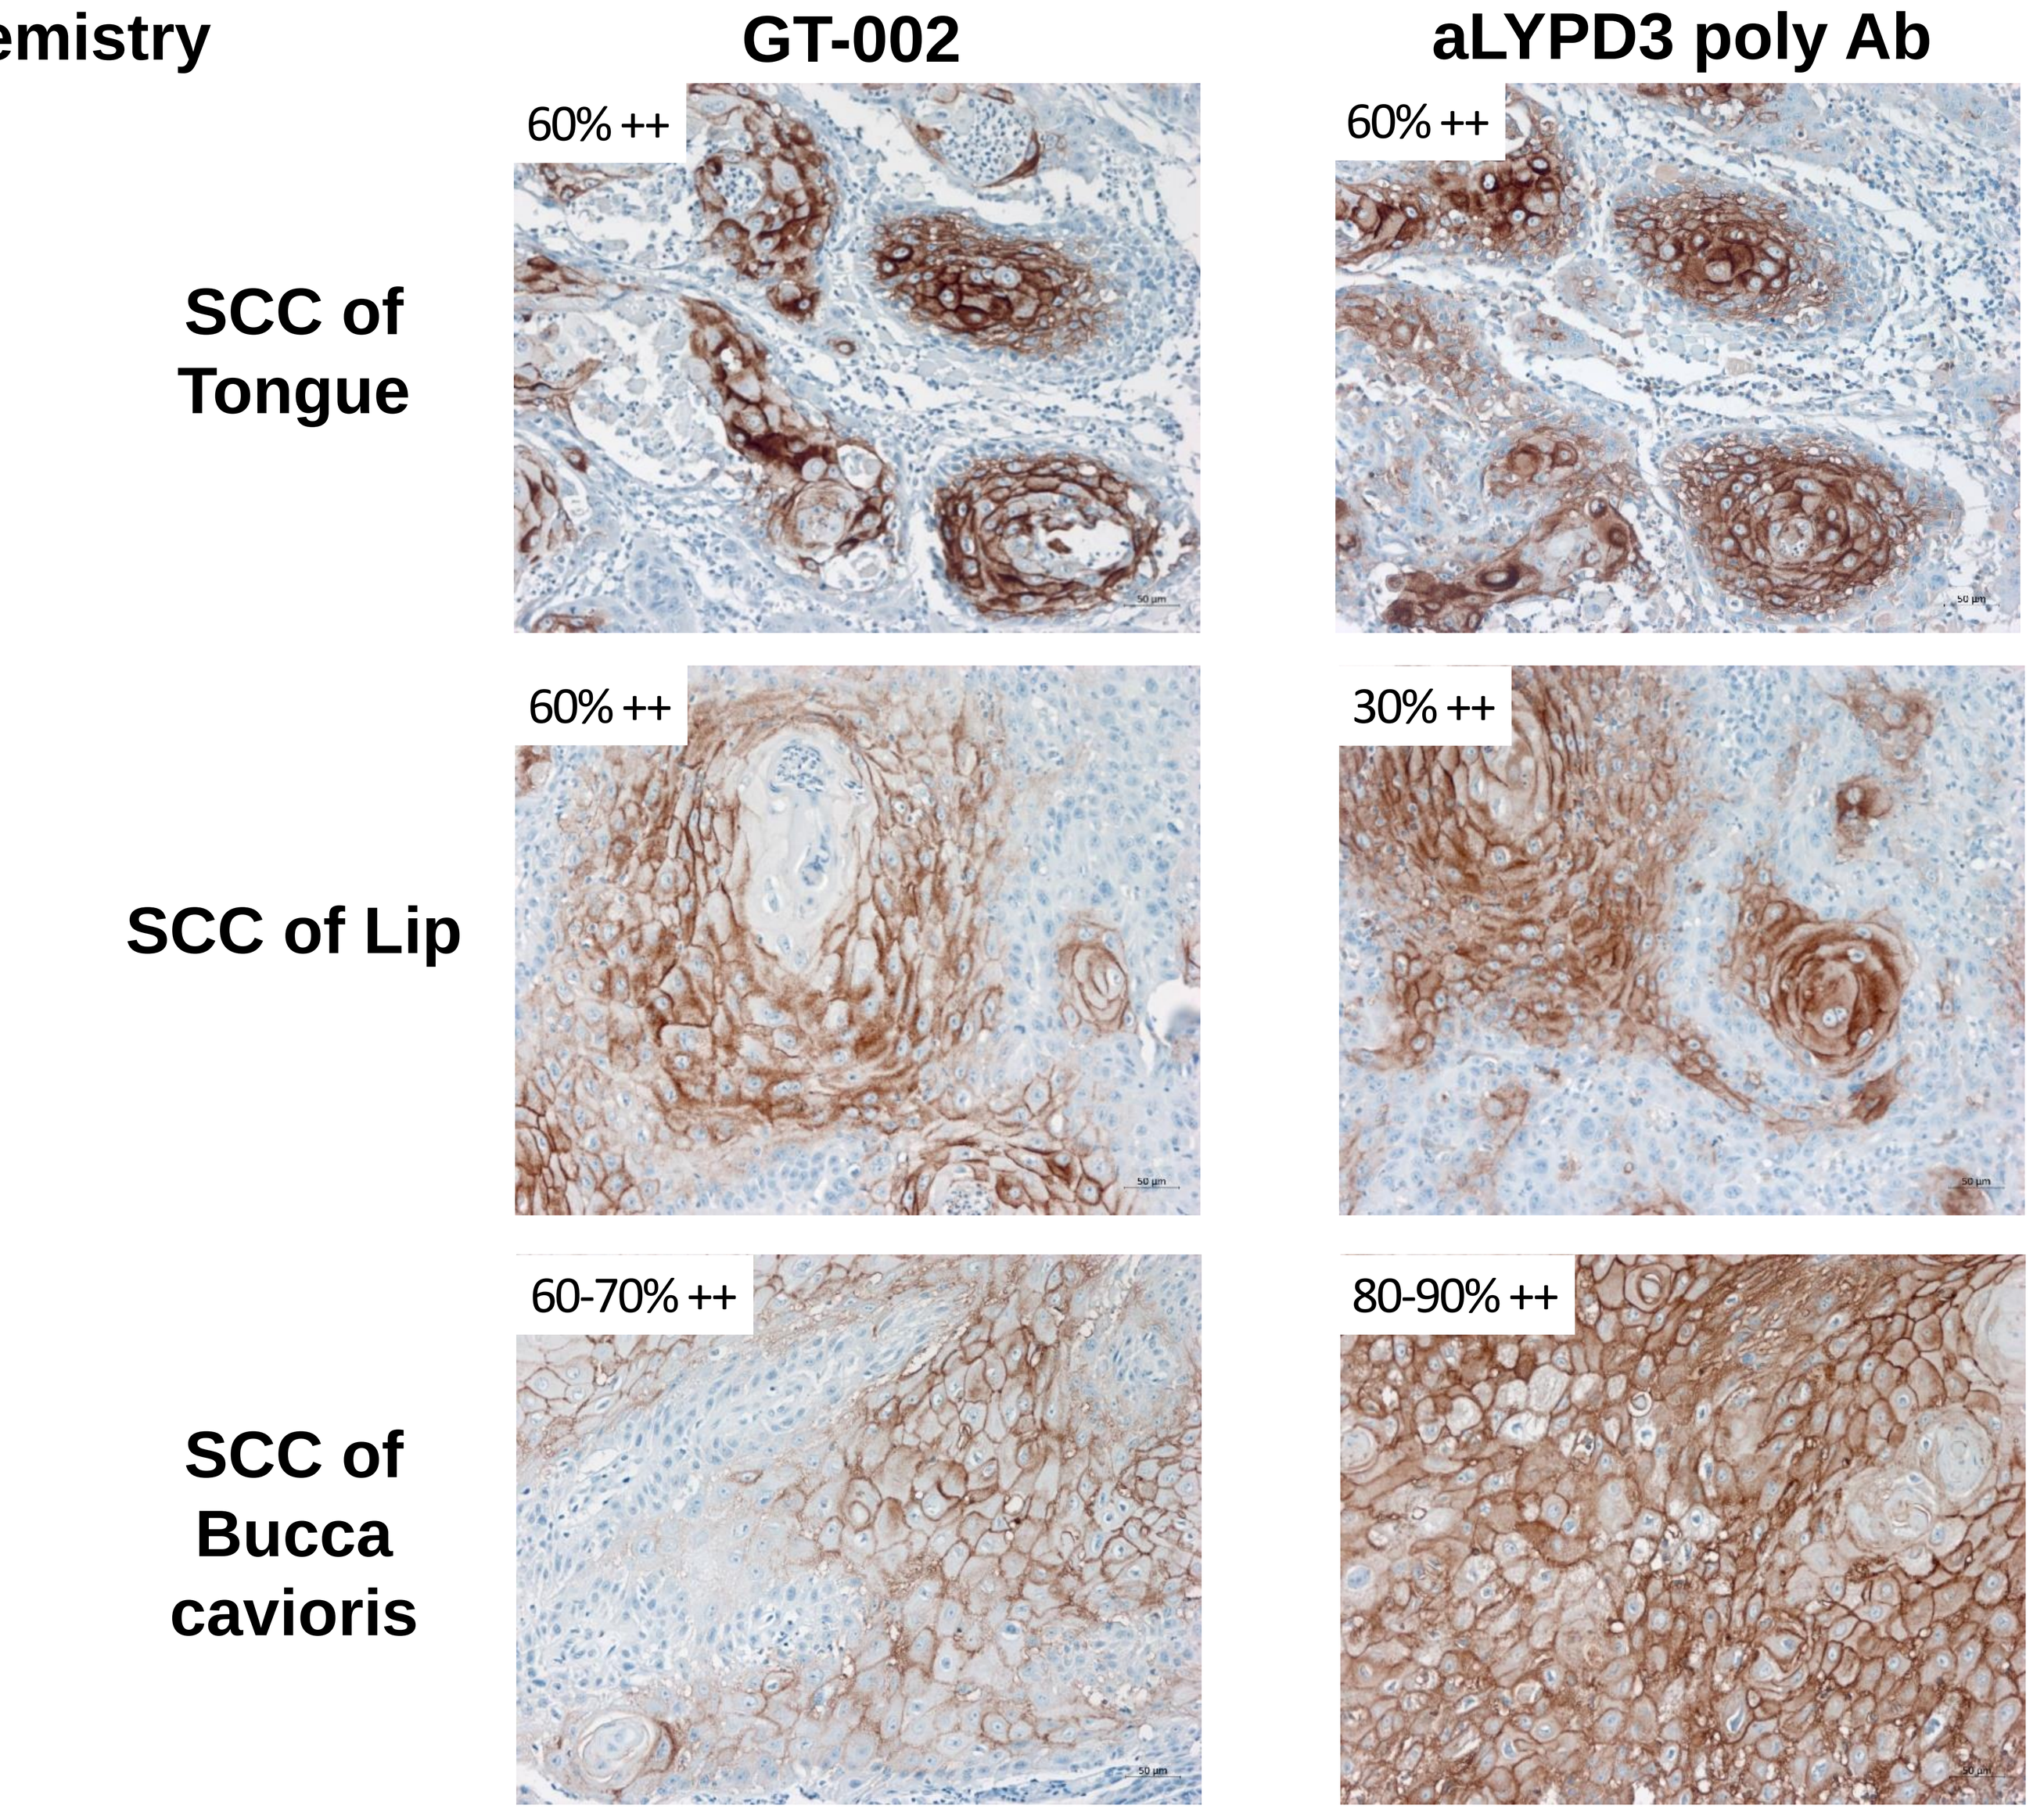
GT-002
aLYPD3 poly Ab
SCC of Tongue
60% ++
60% ++
SCC of Lip
60% ++
30% ++
SCC of Bucca cavioris
60-70% ++
80-90% ++

	Negative #	Negative %	<10% #	<10% %	10-50% #	10-50% %	51-100% #	51-100% %	Total #Cases	Total %pos
Anal area/rectum	2	**20.0**	1	**10.0**	4	**40.0**	3	**30.0**	10	**80.0**
Oral cavity	6	**46.2**	3	**23.1**	2	**15.4**	2	**15.4**	13	**53.8**
Lip	9	**32.1**	3	**10,7**	11	**39.3**	5	**17.9**	28	**67.9**
Bucca cavioris	4	**44.4**	1	**11.1**	3	**33.3**	1	**11.1**	9	**55.6**
Vulva	0	**0.0**	2	**40.0**	2	**40.0**	1	**20.0**	5	**100.0**
Nose	5	**50.0**	1	**10.0**	4	**40.0**	0	**0.0**	10	**50.0**
Penis	2	**40.0**	1	**20.0**	2	**40.0**	0	**0.0**	5	**60.0**
Epiglottis	2	**40.0**	2	**40.0**	1	**20.0**	0	**0.0**	5	**60.0**
Tongue	38	**51.4**	12	**16.2**	16	**21.6**	8	**10.8**	74	**48.6**
Skin	34	**69.4**	4	**8.2**	8	**16.3**	3	**6.1**	49	**30.6**

- GT-002 binds to a similar number of patients and tumor areas as the aLYPD3 poly Ab
- GT-002 shows strongest staining in SCC of anal area/rectum, oral cavity, lip, bucca cavioris, nose, penis, vulva, epiglottis, tongue, and skin

Conclusion

- **GT-002** is an **anti-LYPD3 antibody** that targets neither glycan nor LYPD3 protein alone, but only a **highly tumor-specific protein/carbohydrate (TF) combined epitope of LYPD3**
- GT-002 stains cancer tissue of various indications while showing drastically reduced off-tumor binding towards healthy LYPD3-positive tissues
- We believe that this highly specific antibody is exceptionally **suitable for potent therapies like ADC, radio-conjugates and CARs**
- **Our approach is suitable to generate antibodies targeting tumor-associated protein/carbohydrate combined epitopes on glycoproteins (please see poster 1346)**

References

1) Pinho SS, Reis CA. Glycosylation in cancer: mechanisms and clinical implications. Nat Rev Cancer. 2015 Sep;15(9):540-55. doi: 10.1038/nrc3982. Epub 2015 Aug 20. PMID: 26289314.
2) Kudelka MR, Ju T, Heimburg-Molinaro J, Cummings RD. Simple sugars to complex disease--mucin-type O-glycans in cancer. Adv Cancer Res. 2015;126:53-135. doi: 10.1016/bs.acr.2014.11.002. Epub 2015 Feb 7. PMID: 25727146; PMCID: PMC5812724.
3) Hansen LV, Gårdsvoll H, Nielsen BS, Lund LR, Danø K, Jensen ON, Ploug M. Structural analysis and tissue localization of human C4.4A: a protein homologue of the urokinase receptor. Biochem J. 2004 Jun 15;380(Pt 3):845-57. doi: 10.1042/BJ20031478. PMID: 15012588; PMCID: PMC1224211.
4) Würfel J, Seiter S, Stassar M, Claas A, Kläs R, Rösel M, Marhaba R, Savelyeva L, Schwab M, Matzku S, Zöller M. Cloning of the human homologue of the metastasis-associated rat C4.4A. Gene. 2001 Jan 10;262(1-2):35-41. doi: 10.1016/s0378-1119(00)00515-1. PMID: 11179665.

We aim to continuously expand our collaborations with industry partners and academic centers to further exploit the unique potential of our technology.
For further discussion please contact business.development@glycotope.com or visit our webpage https://www.glycotope.com/contact/