GT-001 - anti-Lewis Y antibody with superior fine-specificity and reduced off-target binding

Patrik Kehler, Anika Jäkel, Timo Lischke, Lisa Weiß, Christoph Goletz, Evelyn Hartung, Anke Flechner, Sven Bahrke, Johanna Gellert, Antje Danielczyk

Patrik.Kehler@glycotope.com; Glycotope, Berlin, Germany

GLYCOTOPE

Introduction

- The Lewis Y (CD174) carbohydrate antigen is widely expressed in primary and metastatic epithelial tumors like colon, lung, ovarian, and breast.
- Targeting Lewis Y for cancer therapy was pursued before, however, other Lewis Y antibodies tested in clinical trials showed cross-reactivity to related carbohydrate structures expressed on blood cells and mostly failed for efficacy and/or safety reasons [1, 2, 3, 4, 5].

We have developed a humanized antibody (GT-001) that shows superior fine specificity and higher affinity compared to clinically tested anti-Lewis Y antibodies BR96 and h3S193.

Target Background:

- The Lewis Y carbohydrate antigen is a difucosylated tetrasaccharide found on type 2 blood group oligosaccharides of glycolipids and glycoproteins.
- Although Lewis Y is mostly expressed throughout fetal development and gradually lost after birth, tumors have been shown to overexpress Lewis Y making it a good target for mAb therapy.

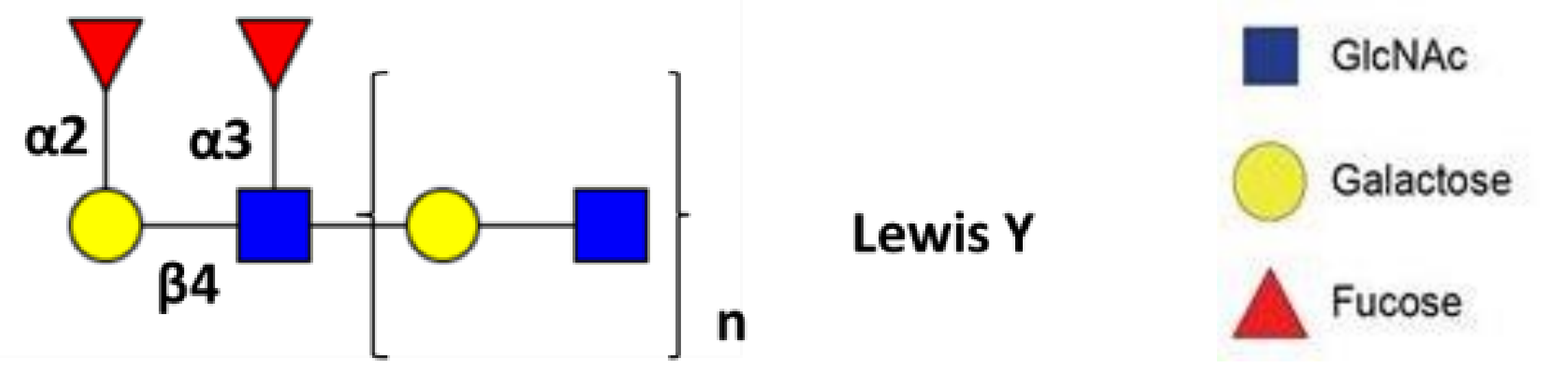

α2
α3
β4
n
Lewis Y
GlcNAc
Galactose
Fucose

Results: Specificity & Affinity

Specificity Screening (ELISA): Binding of GT-001, BR96 and h3S193 to selected PAA conjugates

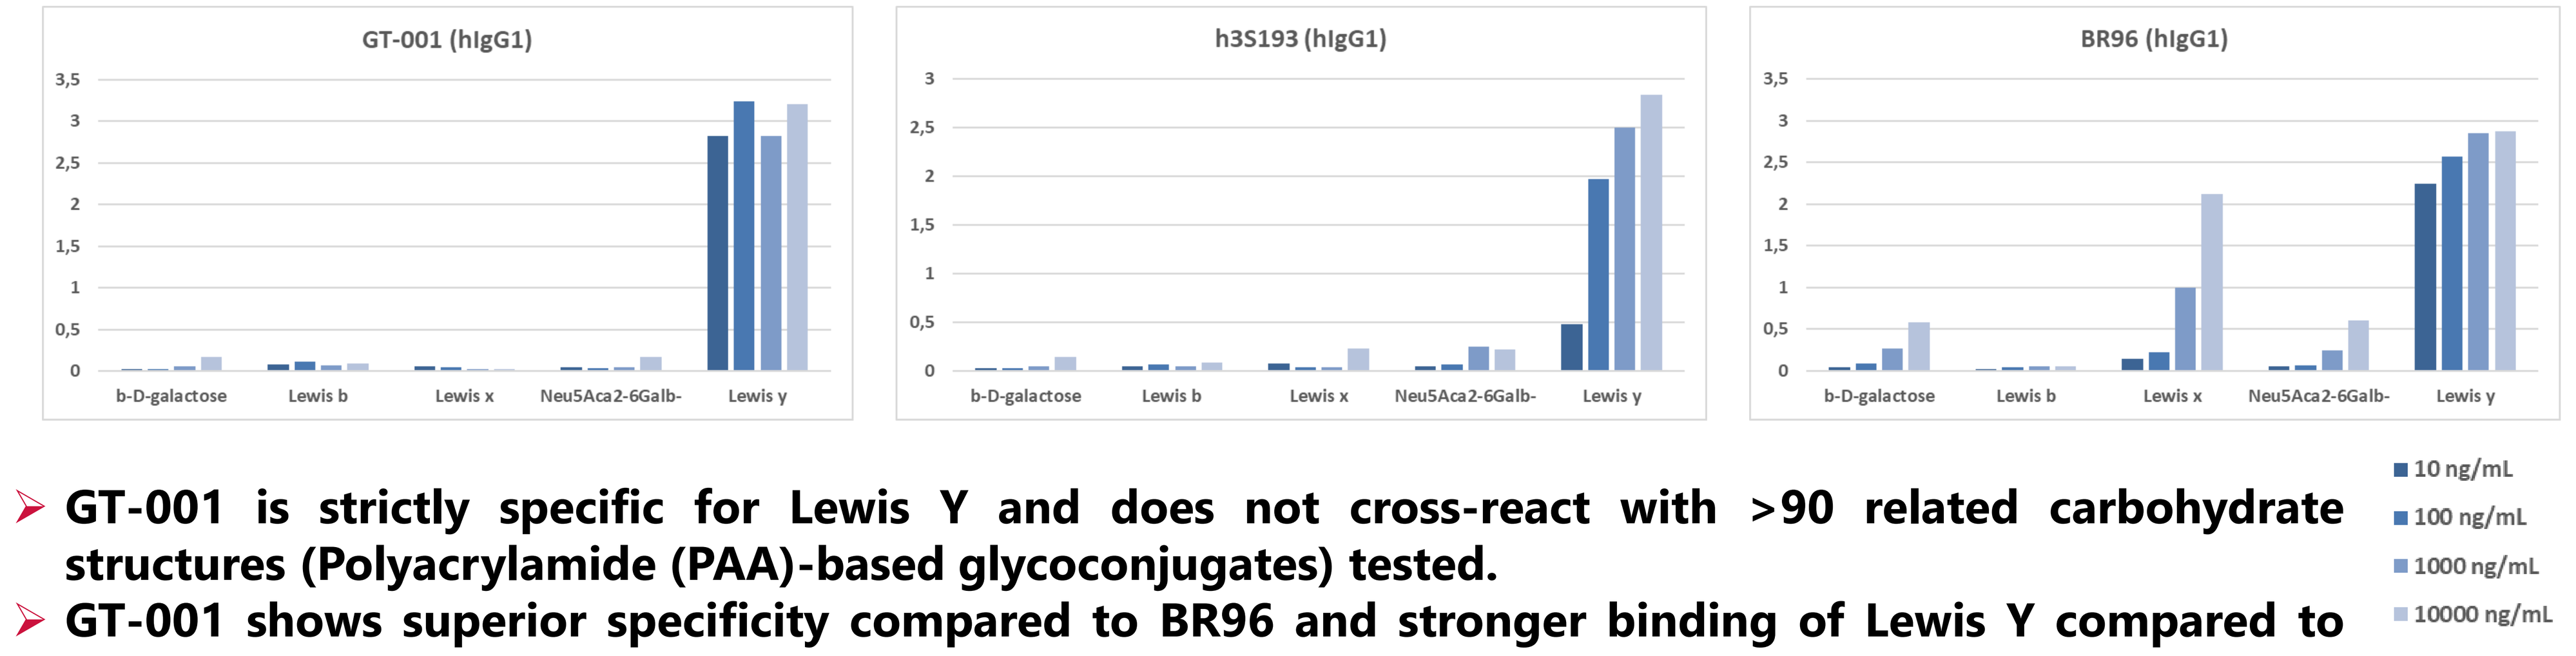

GT-001 (hIgG1)
h3S193 (hIgG1)
BR96 (hIgG1)
b-D-galactose
Lewis b
Lewis x
NeuSAca2-6Galb-
Lewis y
10 ng/mL
100 ng/mL
1000 ng/mL
10000 ng/mL

- ➢ **GT-001 is strictly specific for Lewis Y and does not cross-react with >90 related carbohydrate structures (Polyacrylamide (PAA)-based glycoconjugates) tested.**
- ➢ **GT-001 shows superior specificity compared to BR96 and stronger binding of Lewis Y compared to h3S193.**

Affinity Measurement (switchSENSE® technology (DRX2, Dynamic Biosensors)): Affinity of GT-001, BR96 and h3S193 to Lewis Y PAA-conjugate

Antibody	Ligand	kON [M-1s-1]	kOFF [s-1]	KD [nmol/L]
GT-001	**Lewis Y**	4.41E+05	9.16E-03	**20.77**
BR96	**Lewis Y**	9.46E+05	5.41E-02	**57.19**
h3S193	**Lewis Y**	indeterminable	indeterminable	indeterminable

- ➢ **In a dynamic system GT-001 shows clearly higher binding affinity for Lewis Y compared to BR96 and h3S193 (affinity of h3S193 for LeY-PAA indeterminable).**

GT-001 is available for partnering or co-development. For further discussion please contact business.development@glycotope.com or visit our webpage https://www.glycotope.com/contact/

Results: Blood Cell & Tumor Binding

Blood Cell Binding (FCM): Binding of GT-001, BR96 and h3S193 to human blood cells

PBMCs and granulocytes were isolated from whole blood of six healthy donors and blood cell binding was analyzed

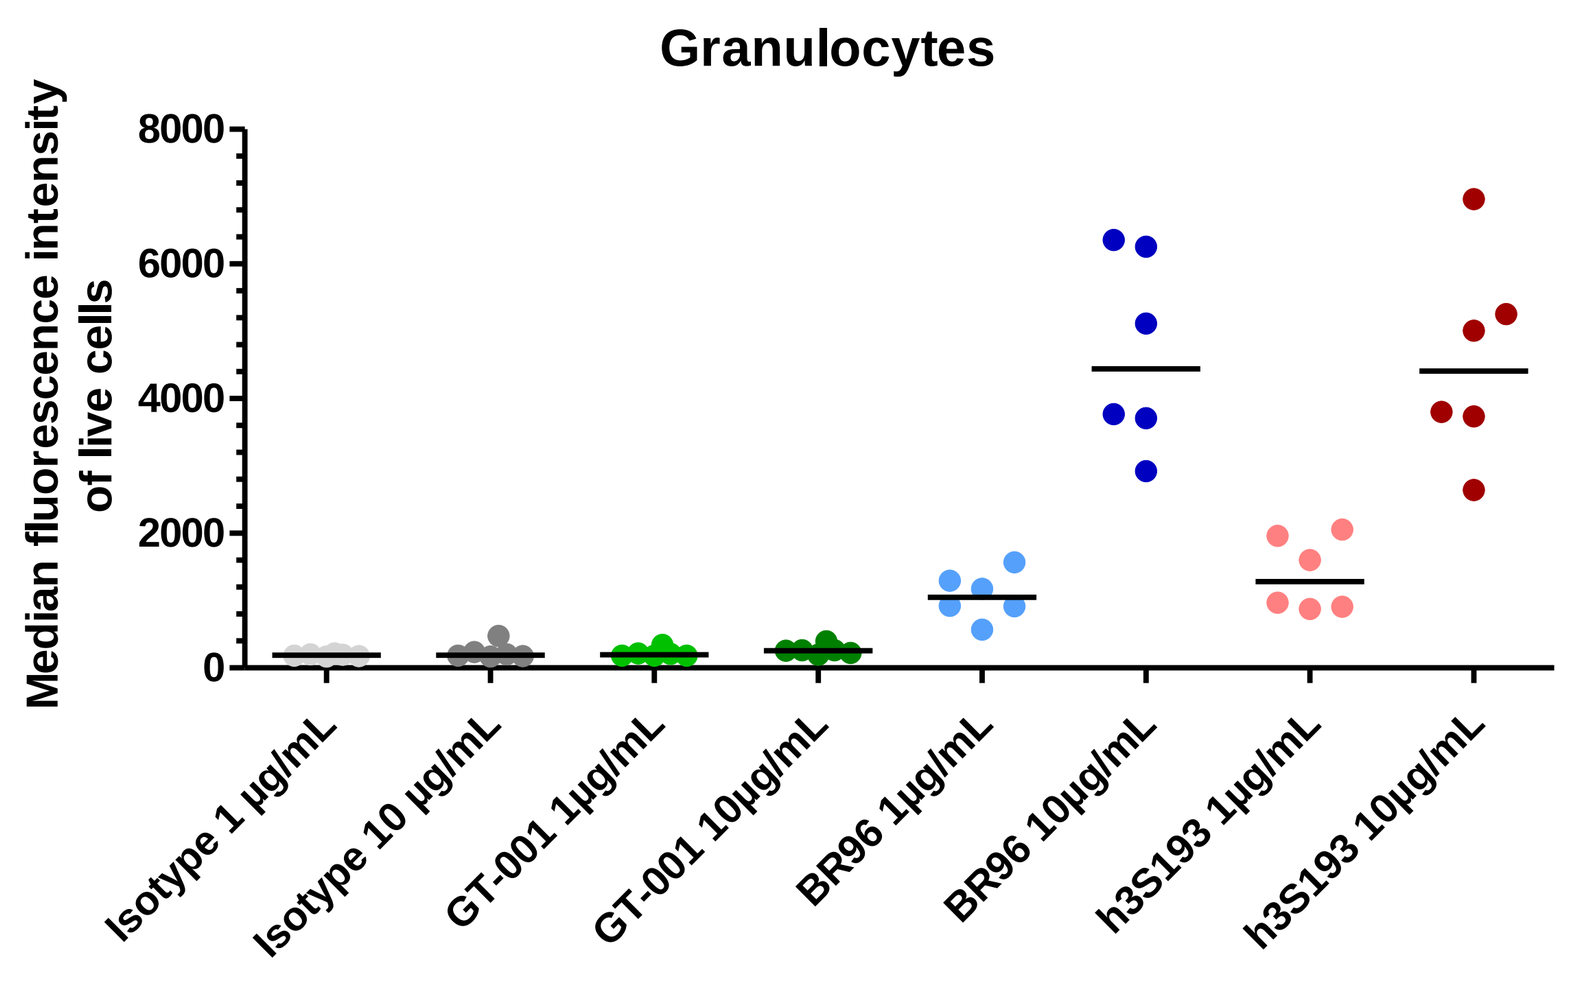

Granulocytes
Median fluorescence intensity of live cells
Isotype 1 µg/mL
Isotype 10 µg/mL
GT-001 1µg/mL
GT-001 10µg/mL
BR96 1µg/mL
BR96 10µg/mL
h3S193 1µg/mL
h3S193 10µg/mL

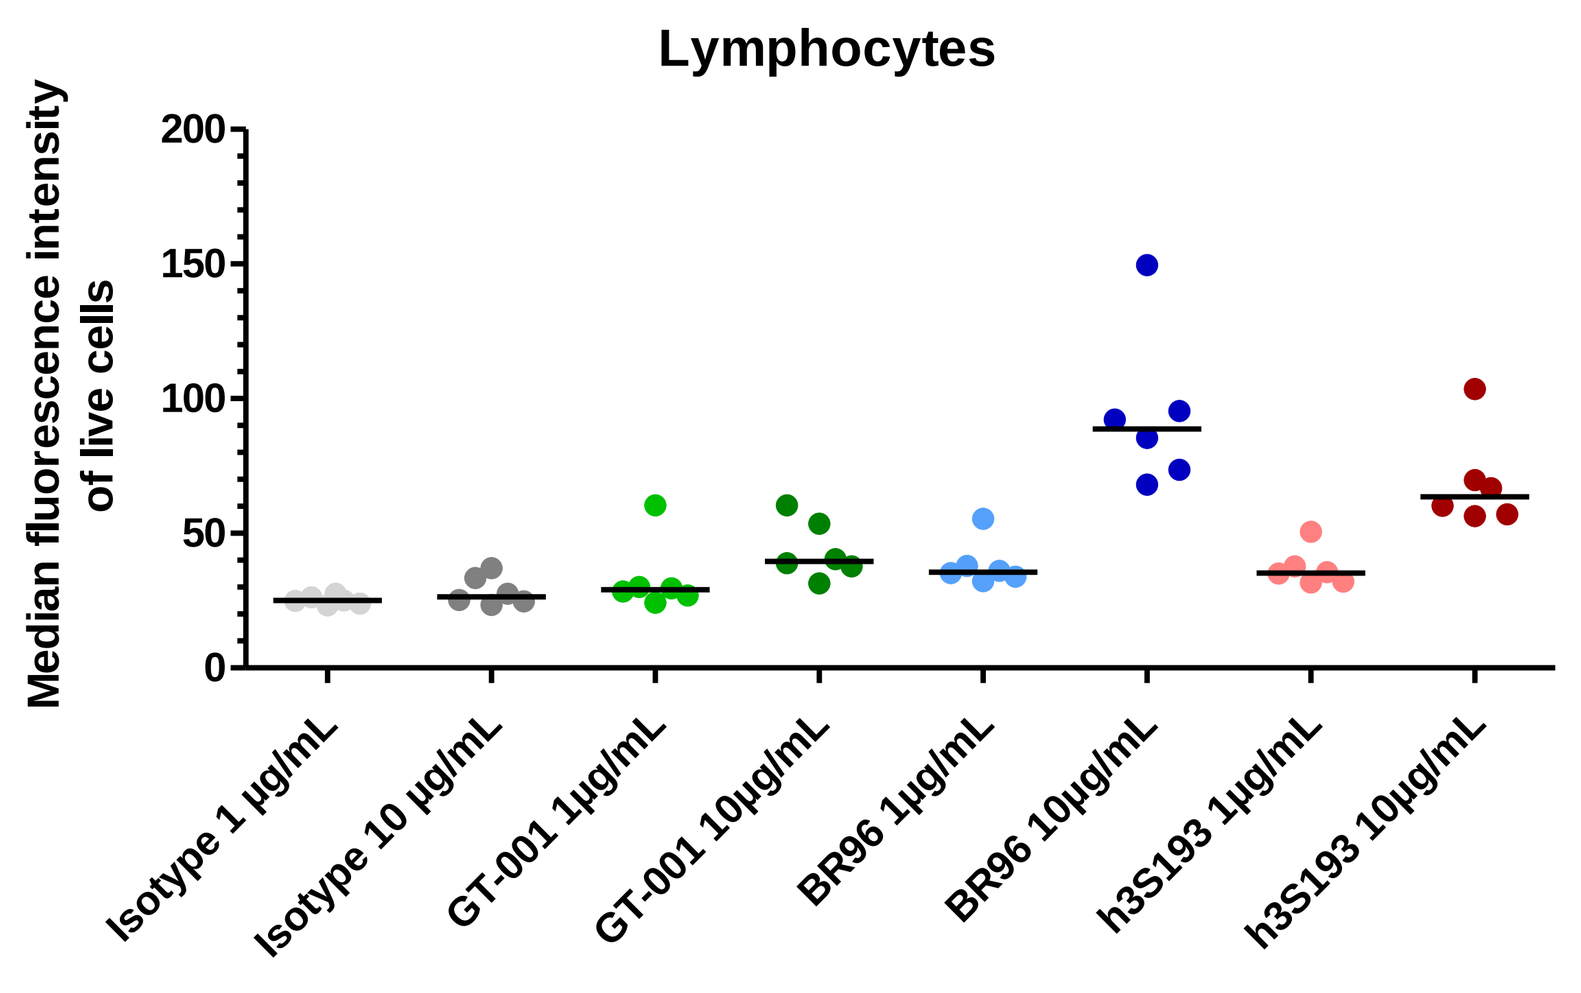

Lymphocytes
Median fluorescence intensity of live cells
Isotype 1 µg/mL
Isotype 10 µg/mL
GT-001 1µg/mL
GT-001 10µg/mL
BR96 1µg/mL
BR96 10µg/mL
h3S193 1µg/mL
h3S193 10µg/mL

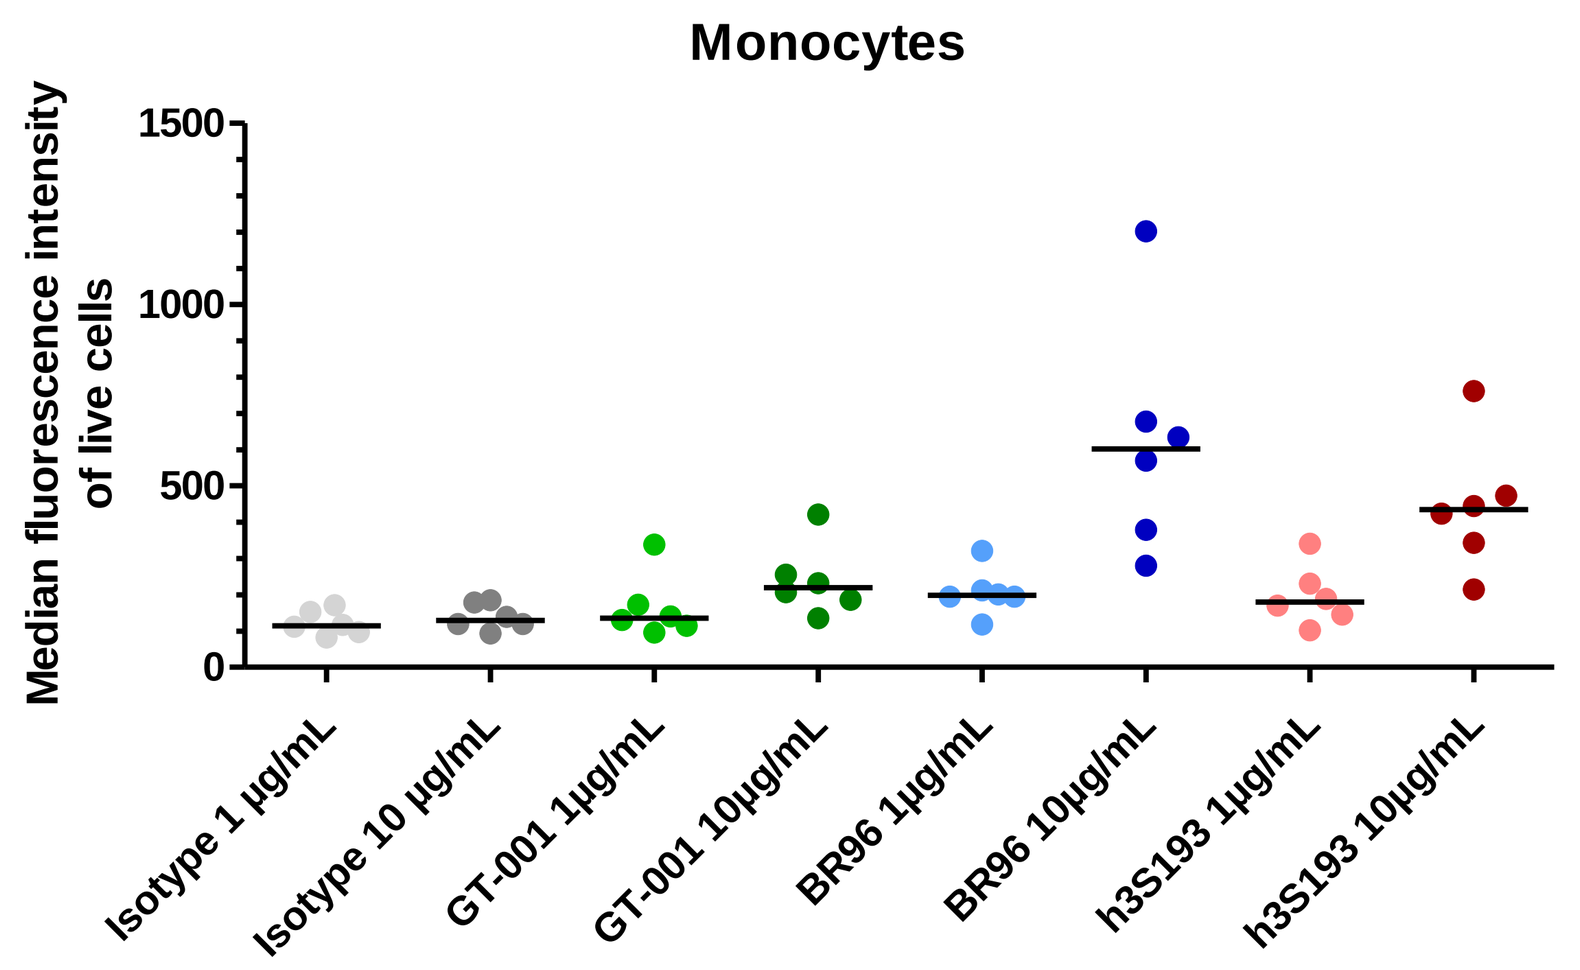

Monocytes
Median fluorescence intensity of live cells
Isotype 1 µg/mL
Isotype 10 µg/mL
GT-001 1µg/mL
GT-001 10µg/mL
BR96 1µg/mL
BR96 10µg/mL
h3S193 1µg/mL
h3S193 10µg/mL

- ➢ **GT-001 does not bind to granulocytes and, depending on the donor, binds only very weakly to monocytes and lymphocytes.**
- ➢ **In contrast, the two comparators BR96 and h3S193 show significantly stronger binding to granulocytes, monocytes and lymphocytes.**
- ➢ **BR96 and h3S193 do also bind to different immune cell progenitors derived from Bone Marrow Mononuclear Cells (BMMCs) (data not shown).**

Binding of GT-001 to tumor tissues (IHC): Binding of GT-001 to different cancer indications was analyzed by immunohistochemistry (staining of tumor TMA's at 6.5 µg/ml)

		NSCLC	CRC	BRC	HNC	SCLC	OvCa	HCC
Negative	(IRS = 0)	10	10	92	22	24	59	58
Weakly positive	(IRS = 1-3)	12	9	48	23	18	27	7
Moderately positive	(IRS = 4-8)	11	11	37	21	3	14	5
Strongly positive	(IRS = 9-12)	2	5	5	3	0	0	0
Positive cases / total cases		**25/35**	**25/35**	**90/182**	**47/69**	**21/45**	**41/100**	**12/70**
% positive cases		**71.4%**	**71.4%**	**49.5%**	**68.1%**	**46.7%**	**41%**	**17.1%**

IRS score (range 0-12); calculated by staining intensity (range 0-3) x percentage of stained cells (range 0-4).

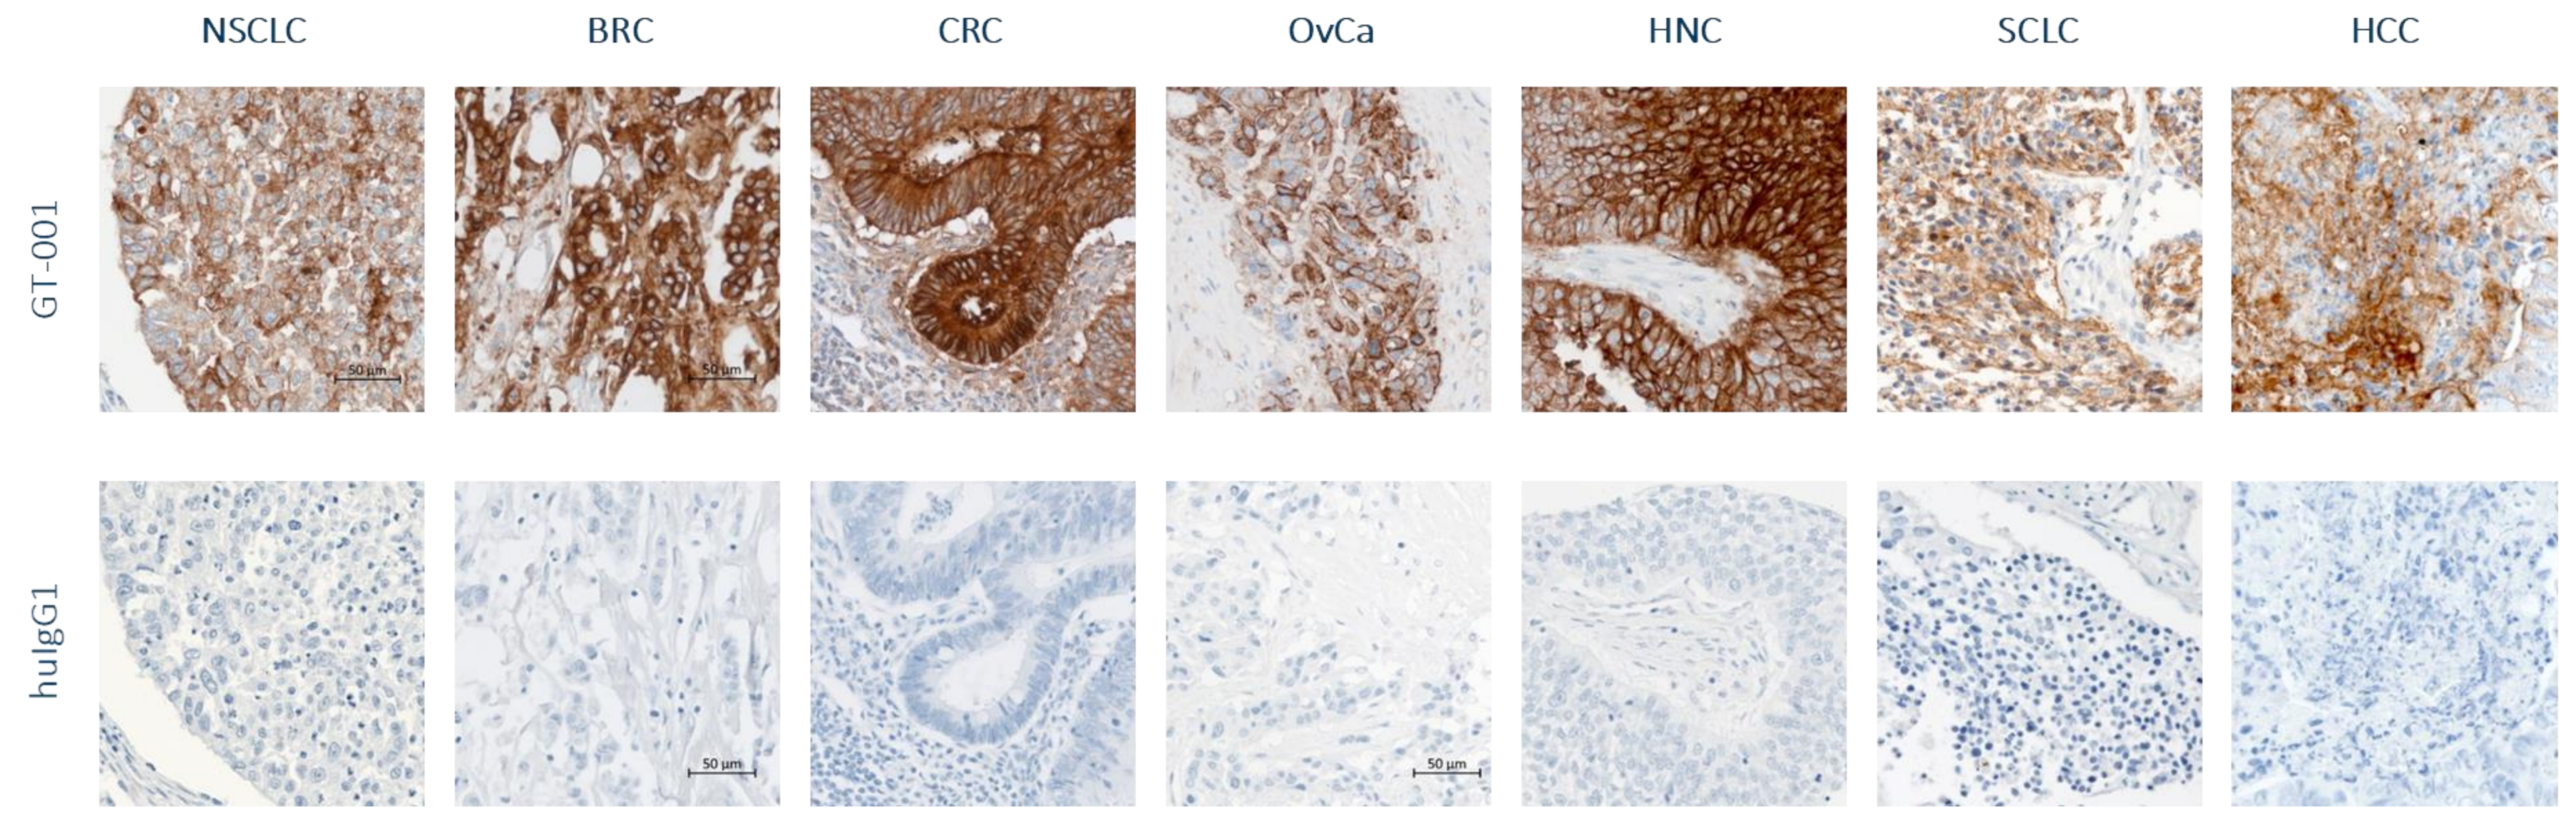

NSCLC
BRC
CRC
OvCa
HNC
SCLC
HCC
GT-001
huIgG1

- ➢ **GT-001 shows apolar membranous staining in various cancer indications and stains tumor tissue of BRC, NSCLC, CRC, HNC, SCLC and OvCa at a high percentage of cases.**

Results: Internalization & Proliferation Inhibition

Internalization & Proliferation Inhibition: Proliferation inhibition (4d) of GT-001 coupled to ProtG-MMAE as surrogate ADC is shown

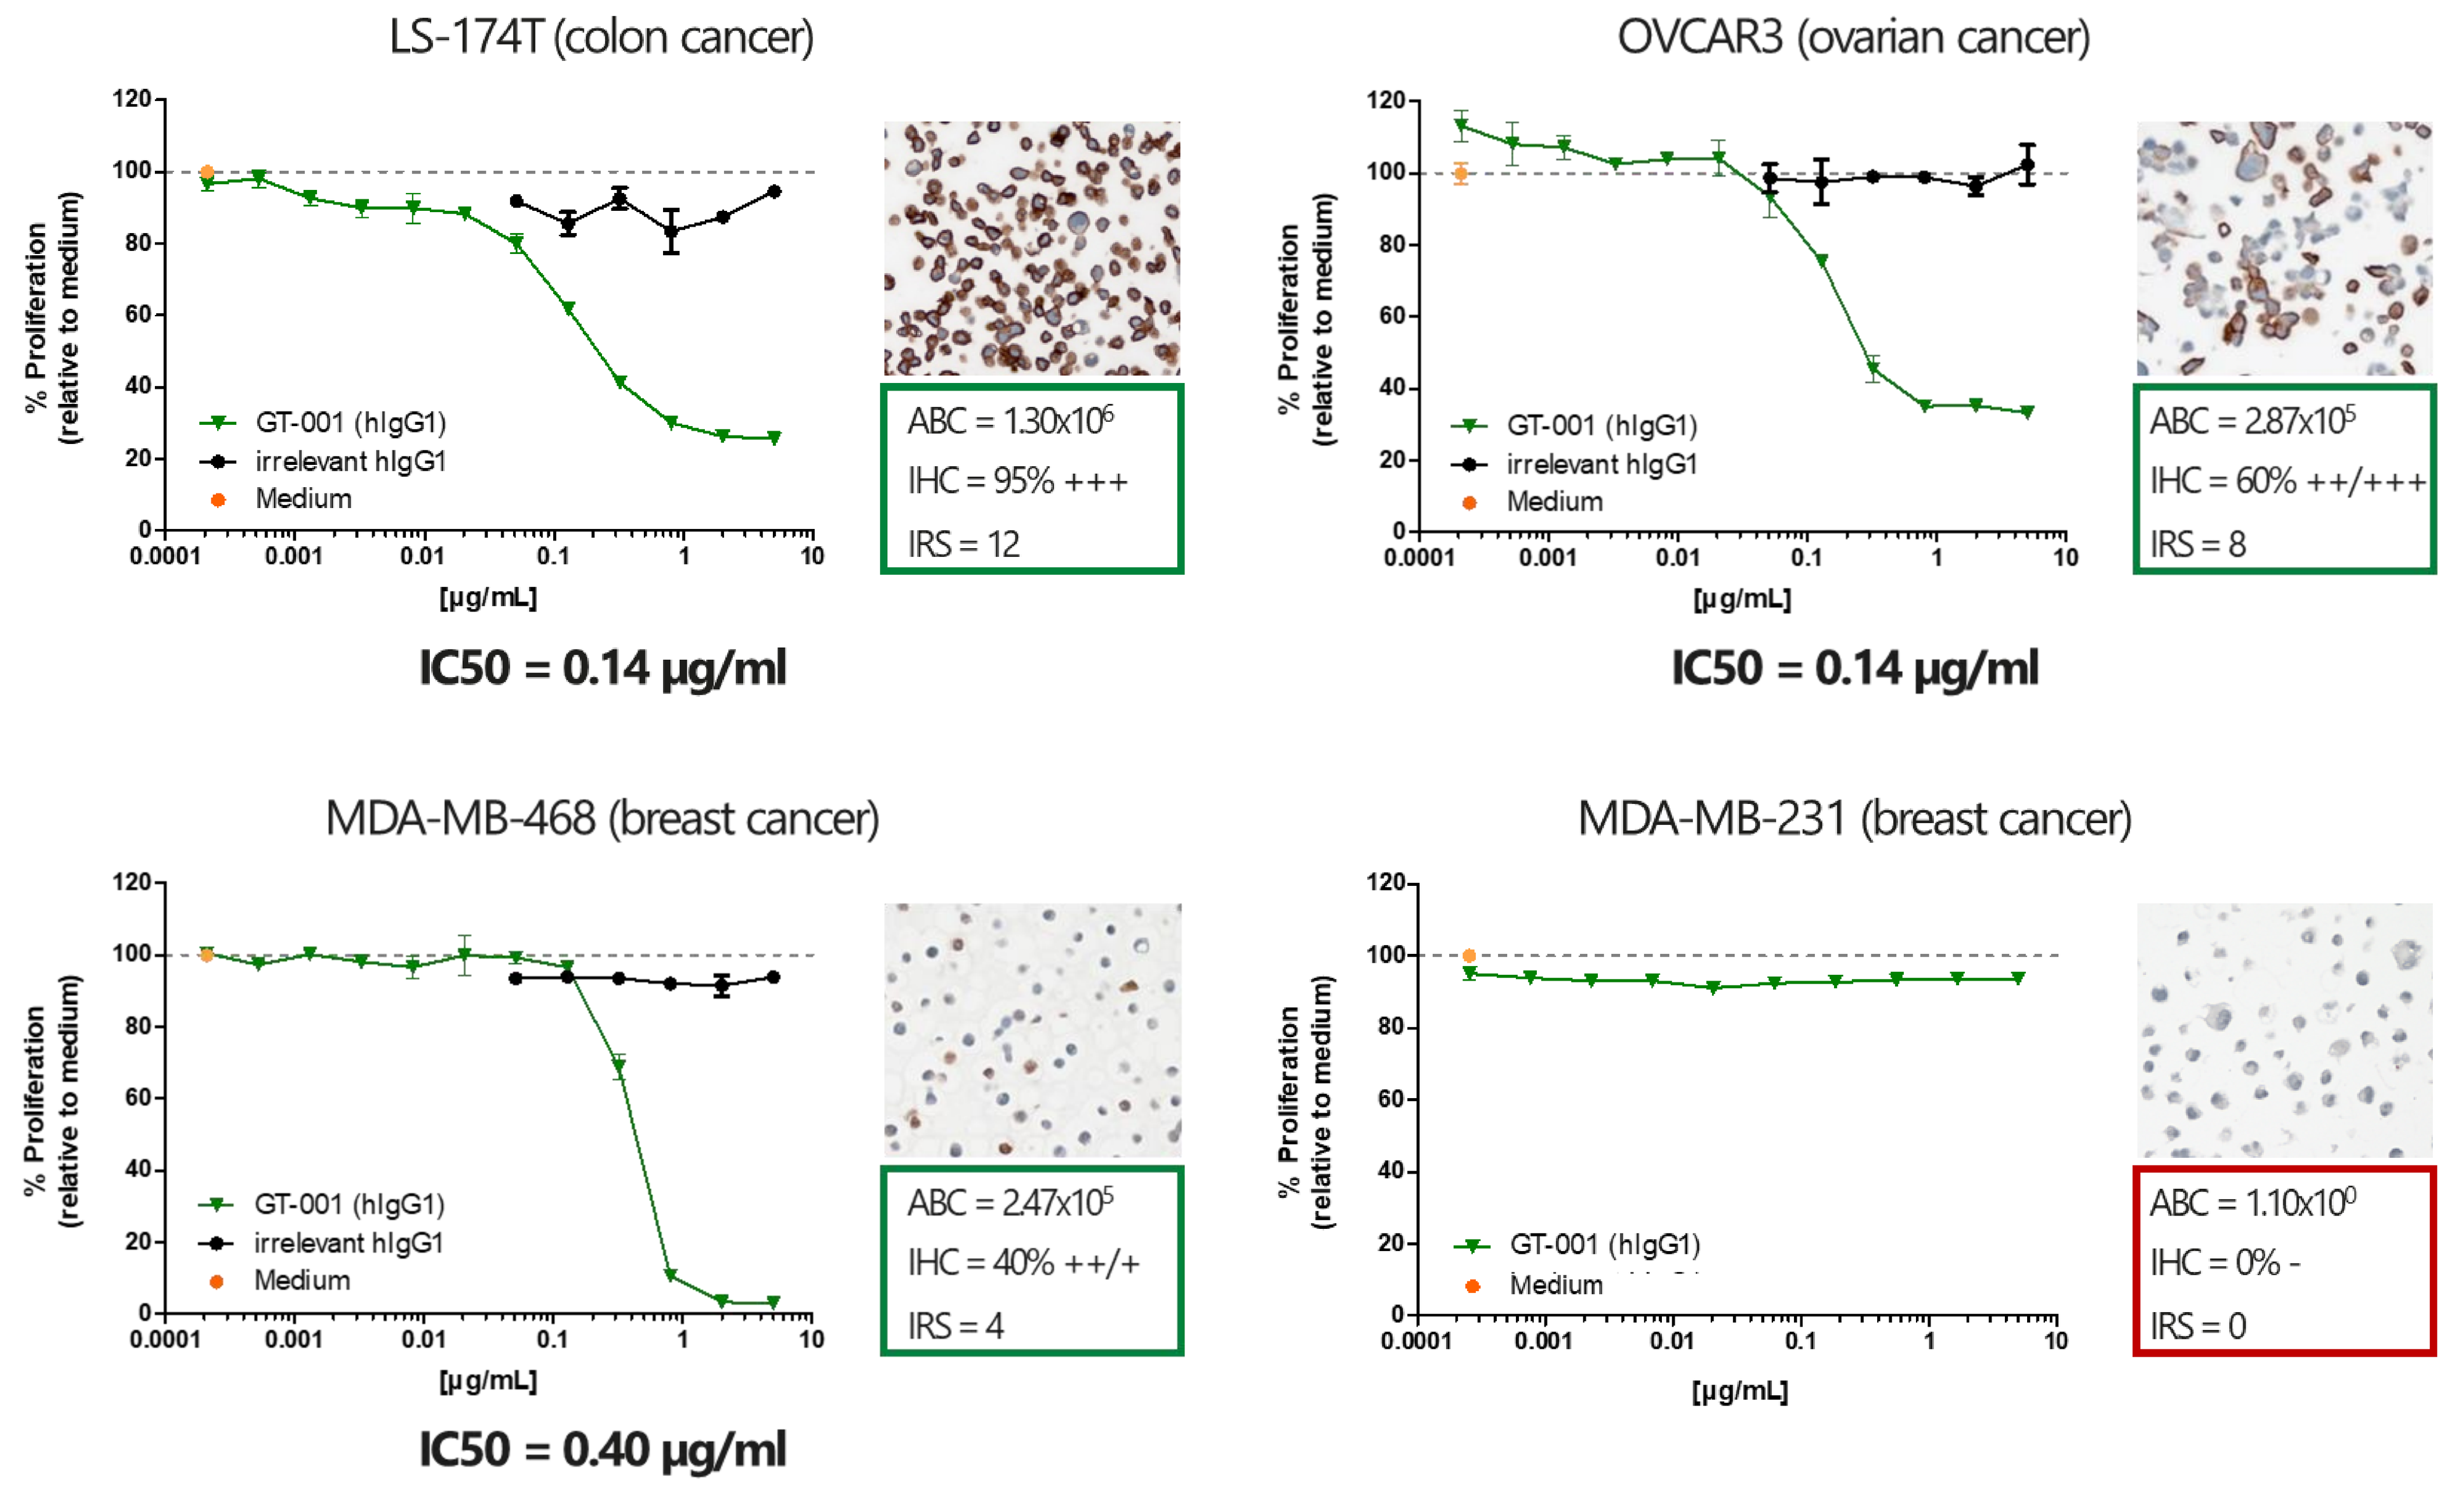

LS-174T (colon cancer)
% Proliferation (relative to medium)
GT-001 (hIgG1)
irrelevant hIgG1
Medium
[µg/mL]
ABC = 1.30x10⁶
IHC = 95% +++
IRS = 12
IC50 = 0.14 µg/ml
OVCAR3 (ovarian cancer)
ABC = 2.87x10⁵
IHC = 60% ++/+++
IRS = 8
IC50 = 0.14 µg/ml
MDA-MB-468 (breast cancer)
ABC = 2.47x10⁵
IHC = 40% ++/+
IRS = 4
IC50 = 0.40 µg/ml
MDA-MB-231 (breast cancer)
ABC = 1.10x10⁰
IHC = 0% -
IRS = 0

- ➢ **GT-001 ADC surrogate induces dose-dependent proliferation inhibition of several Lewis Y-positive cell lines of different origin indicating effective internalization.**

ABC (flow cytometry) = Antibody binding sites per cell

Conclusion

GT-001 - anti-Lewis Y antibody with superior fine-specificity:

- ➢ Lewis Y is expressed on many epithelial tumor indications of high medical need.
- ➢ Several approaches targeting Lewis Y for cancer therapy were pursued before. However, other LeY mAbs like BR96 and h3S193 tested in clinical trials so far were cross-reactive and mostly failed for efficacy and/or safety reasons.
- ➢ We have developed a humanized antibody that shows superior fine specificity and higher affinity compared to clinically tested anti-Lewis Y antibodies BR96 and h3S193.
- ➢ Due to the superior fine-specificity, GT-001 shows no/reduced binding of healthy leukocytes potentially reducing side effects.
- ➢ Expression of Lewis Y in tumors of different origin (e.g. breast, colon, lung, ovarian and head and neck) was confirmed via IHC.
- ➢ Its strong target binding and internalization properties make GT-001 an ideal candidate for ADC development.

Literature:

(1) Ajani JA, Kelsen DP, Haller D, Hargraves K, Healey D. A multi-institutional phase II study of BMS-182248-01 (BR96-doxorubicin conjugate) administered every 21 days in patients with advanced gastric adenocarcinoma. Cancer J. 2000;6(2):78-81.
(2) Saleh MN, Sugarman S, Murray J, Ostroff JB, Healey D, Jones D, Daniel CR, LeBherz D, Brewer H, Onetto N, LoBuglio AF. Phase I trial of the anti-Lewis Y drug immunoconjugate BR96-doxorubicin in patients with lewis Y-expressing epithelial tumors. J Clin Oncol. 2000;18(11):2282-92.
(3) Scott AM, Tebbutt N, Lee FT, Cavicchiolo T, Liu Z, Gill S, Poon AM, Hopkins W, Smyth FE, Murone C, MacGregor D, Papenfuss AT, Chappell B, Saunder TH, Brechbiel MW, Davis ID, Murphy R, Chong G, Hoffman EW, Old LJ. A phase I biodistribution and pharmacokinetic trial of humanized monoclonal antibody Hu3s193 in patients with advanced epithelial cancers that express the Lewis-Y antigen. Clin Cancer Res. 2007;13(11):3286-92.
(4) Smaletz O, Diz MD, do Carmo CC, Sabbaga J, Cunha-Junior GF, Azevedo SJ, Maluf FC, Barrios CH, Costa RL, Fontana AG, Madrigal V, Wainstein AJ, Yeda FP, Alves VA, Moro AM, Blasbalg R, Scott AM, Hoffman EW. A phase II trial with anti-Lewis-Y monoclonal antibody (hu3S193) for the treatment of platinum resistant/refractory ovarian, fallopian tube and primary peritoneal carcinoma. Gynecol Oncol. 2015;138(2):272-7.
(5) Zhang S, Zhang HS, Cordon-Cardo C, Reuter VE, Singhal AK, Lloyd KO, Livingston PO. Selection of tumor antigens as targets for immune attack using immunohistochemistry: II. Blood group-related antigens. Int J Cancer. 1997;73(1):50-6.